2009-11-02

Drug delivery on time by Egalet® CHRONO technology

Christensen L.H.†,Nilsson M.S., Hemmingsen P.H., Tygesen P.

Egalet a/s, DK-3500 Værløse, Denmark, †lhc@egalet.com

INTRODUCTION

A major and withstanding challenge within the pharmaceutical industry is formulation of precisely timed therapeutics. These could present an important benefit for both patients and society within indications that have cyclic symptoms. Examples of such indications include asthma, arthritis and Addisons disease [1].

Egalet a/s is a pharma company specialized in modified release. Egalet® technolgies offer controlled release profiles ranging from zero order release to delayed release as well as combinations thereof [2].

The fundament of Egalet® formulations are based on an injection molded polymer system consisting of an erodible matrix partly covered with a water-impermeable, non-erodible shell (Figure 1). The well-defined fixed surface erosion area at both ends of the cylindrical tablet allows a tightly controlled, extended release as well as the possibility to formulate delayed release preparations (Figure 2).

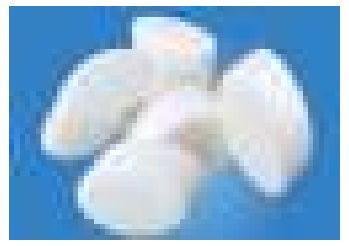

Figure 1. Egalet® tablet

The active substance is dispersed in a matrix partly covered by an essentially non-erodible shell . The drug is released by erosion of the matrix.

MATERIALS AND EXPERIMENTAL METHODS

Several different formulations were tested to find a immediate release formulation for the active matrix, which were suitable for injection molding.

The shell material was made of ethyl cellulose and was injection molded into a basket shape with only one end open.

Production of the active matrix, the delay matrix and the shell was performed on a Haake Minijet II, (Thermo Electron, Karlsruhe, Germany). After injection molding of the three components the finished product was assembled by hand.

Dissolution test of the finished product was performed according to USP apparatus 2 (paddle method) using a 1 N HCl solution, pH 1.2 or a phosphate buffer pH 6.8.

.

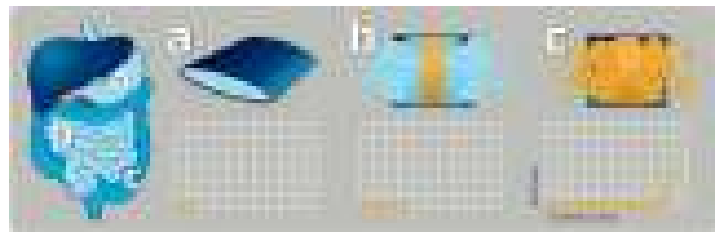

Figure 2. Schematic overview of product design

A: Finished product; B: Delay matrix starts to erode; C: Active matrix starts to erode.

RESULTS AND DISCUSSION

Precise control of the duration of the delay was obtained by controlling the length of the delay matrix (Figure 3). In particular, proportionality was found between the length of the delay matrix and the onset of the release. This supports that erosion is the main release principle for the delay matrix.

In order to minimize the length of the finished product the shell was closed in one end and the product formulated with one delay and one active matrix. The closing of one end results in an increased time from onset of release to end of release of the API compared to a product which is open in both ends. If a burst effect is critical to the product, a shell with both ends open leads to the shortest release time possible.

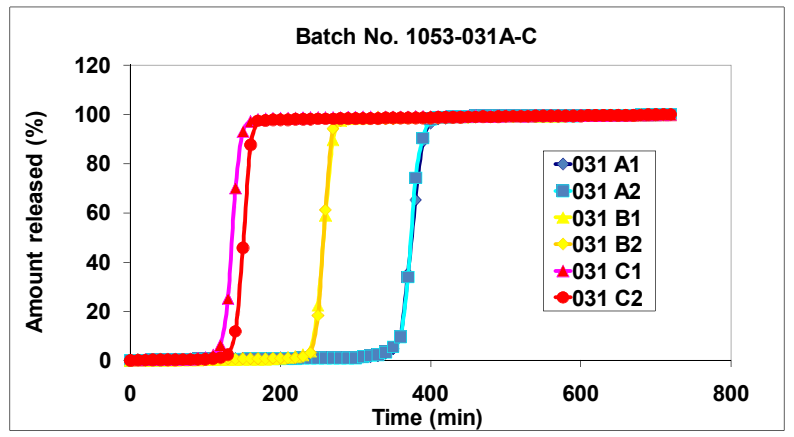
Batch No. 1053-031A-C
Amount released (%)
120
100
80
60
40
20
0
0
200
400
600
800
Time (min)
031 A1
031 A2
031 B1
031 B2
031 C1
031 C2

Figure 3. In vitro dissolution depends on geometry

The time from medication until onset of release is controlled by length of the delay matrix. Red and pink: short, yellow and orange: medium and blue and turquoise: long delay matrix.

The steepness of the release curve is mainly related to the formulation of the active matrix but the formulation of the delay matrix might also contribute to the release profile by diffusion, depending on the excipients used in the delay matrix.

Dissolution has been performed in both pH 6.8 and 1.2. No significant differences were seen neither in delay time nor in the time from onset of release until 100 % released (Figure 4).

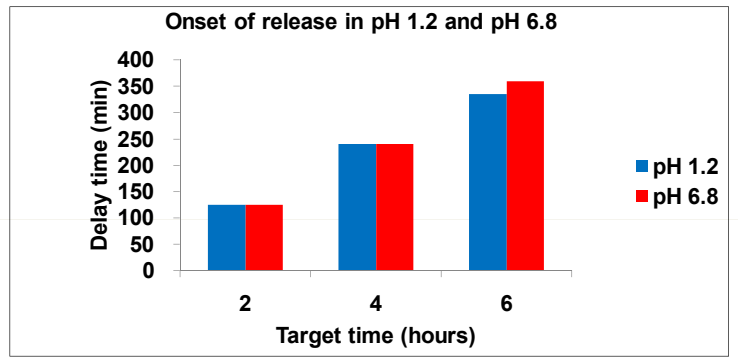
Onset of release in pH 1.2 and pH 6.8
Delay time (min)
400
350
300
250
200
150
100
50
0
2
4
6
Target time (hours)
pH 1.2
pH 6.8

Figure 4. Time until onset of release

Blue: pH 1.2 and Red: pH 6.8 USP dissolution media.

CONCLUSION

It is possible to predict the onset of API release very accurately by the length of the chosen delay matrix entrapping the API. The length of the delay matrix was found to have a linear dependency on the onset of API release.

Many patients suffer the worst symptoms in the morning. Among these are people suffering from allergic rhinitis, rheumatoid arthritis and asthma. For these groups of patients it would be beneficial if the medication worked before standing out of bed in the morning [1], [2].

Egalet® CHRONO technology offers precise and reliable drug delivery to such indications where delayed release is a requirement.

REFERENCES

[1] Bi-Botti C. Youan: "Chronopharmaceutics: Science and Technology for Biological Rhythm-Guided Therapy and Prevention of Diseases."

[2] Neena Washington and Clive G. Wilson: Can Oral Controlled Drug Delivery Meet the Challenges Posed by Chronotherapeutics? Drug Delivery Tech. May 2006, Vol 6, No. 5

1